European Academy of Allergy and Clinical Immunology & World Allergy Organization
World Allergy & Asthma Congress 2013, 22 – 26 June, Milan, Italy

TOLERABILITY AND EFFICACY OF HDM INJECTIVE RUSH IMMUNOTHERAPY WITH MONOMERIC ALLERGOID COMPARED TO SUBLINGUAL ADMINISTRATION IN PATIENTS WITH ALLERGIC RHINITIS

Giulio BRIVIO[1], Maria Assunta BOSCOLO[1], **Enrico COMPALATI**[2]

[1]*Department of Allergy Unit, Hospital of Merate, Italy*

[2]*Department of Allergy And Respiratory Diseases Clinic, University of Genoa, Italy*

Background

Monomeric carbamylated allergoids (monoids) have reduced IgE-binding activity resulting safe by sublingual or subcutaneous administration. They reduce allergic symptoms and the use of drugs. Injections of mite (HDM) and grass pollen monoid are well tolerated following 5 weeks build-up phase. We investigated the safety and efficacy of a treatment course with a rush induction phase.

Method

This open pilot study included 30 mono-sensitized patients receiving monthly for 12 months a preparation of 0.50 mL of 10 BU/mL containing monomeric allergoid extract (50% Dermatophagoides pteronyssinus and 50% Dermatophagoides farinae, adsorbited on Ca-fosfate, starting with a single day induction (0.20 mL plus 0.50 mL after half an hour).
As control 30 patients were treated with HDM sublingual monoid (1000 AU twice a week). Visual analogue score (VAS) was used to evaluate the symptoms of rhinoconjunctivitis at baseline and monthly.
Drug consumption was calculated with a monthly score. Adverse reactions (ARs) to treatment were recorded at each visit.

Results

All patients completed the study. No serious ARs occurred. The rush induction resulted safe. No ARs occurred in 21 patients (70%), 6 patients (30%) referred local mild ARs not requiring interruption. During the maintenance phase the treatment was well tolerated. A progressive improvement in mean monthly VAS observed during winter months ($p<0.001$) without any significant difference between groups throughout the year ($p>0.05$). Drug intake was comparable between groups ($p>0.05$).
The number of doctors and patients with optimal or good judgment of the treatment efficacy and tolerability was similar.

Conclusion

The safety of monoid appears confirmed also through injective route with rush induction scheme. The improved health condition and drug usage were comparable with those achieved by patients treated with monoid administered sublingually.

Figure 1. Results - Safety

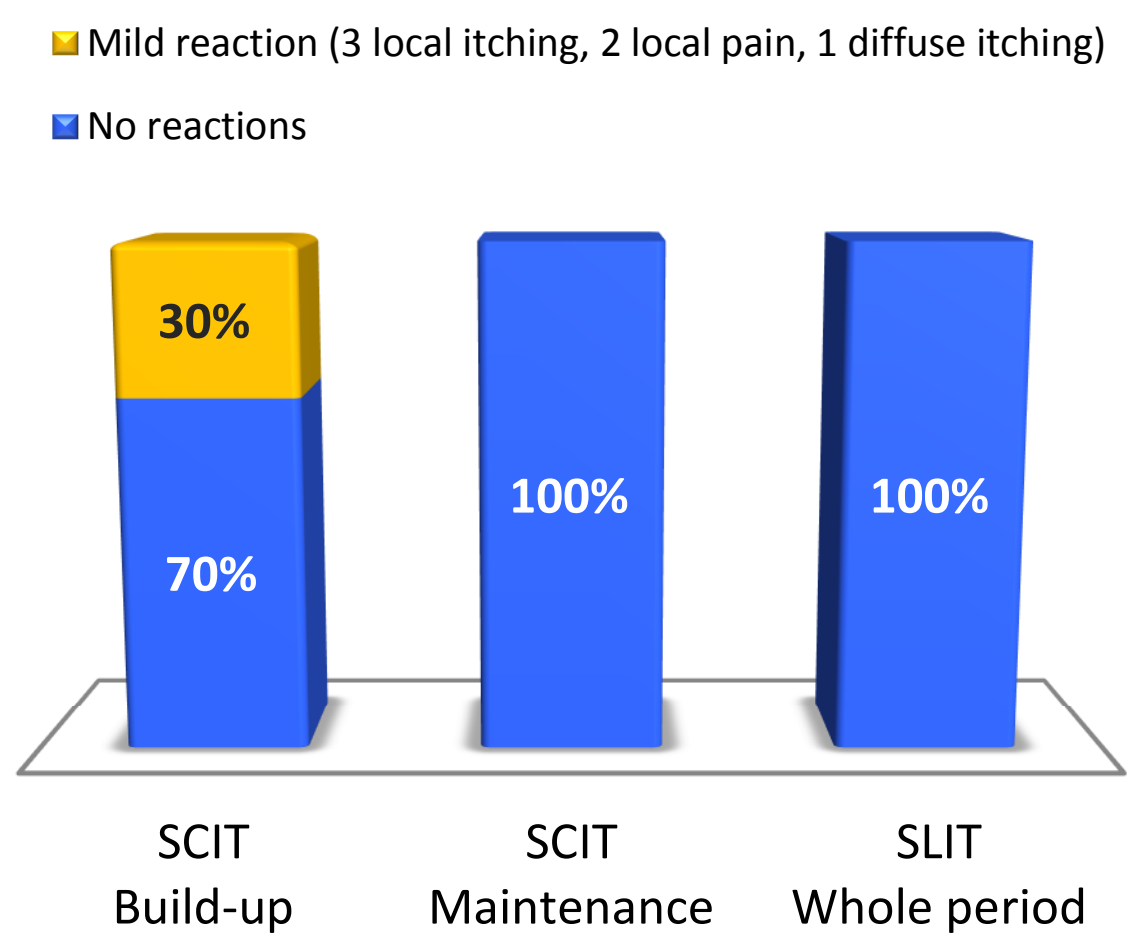
Mild reaction (3 local itching, 2 local pain, 1 diffuse itching)
No reactions
30%
70%
100%
100%
SCIT Build-up
SCIT Maintenance
SLIT Whole period

Figure 2. Results - Efficacy

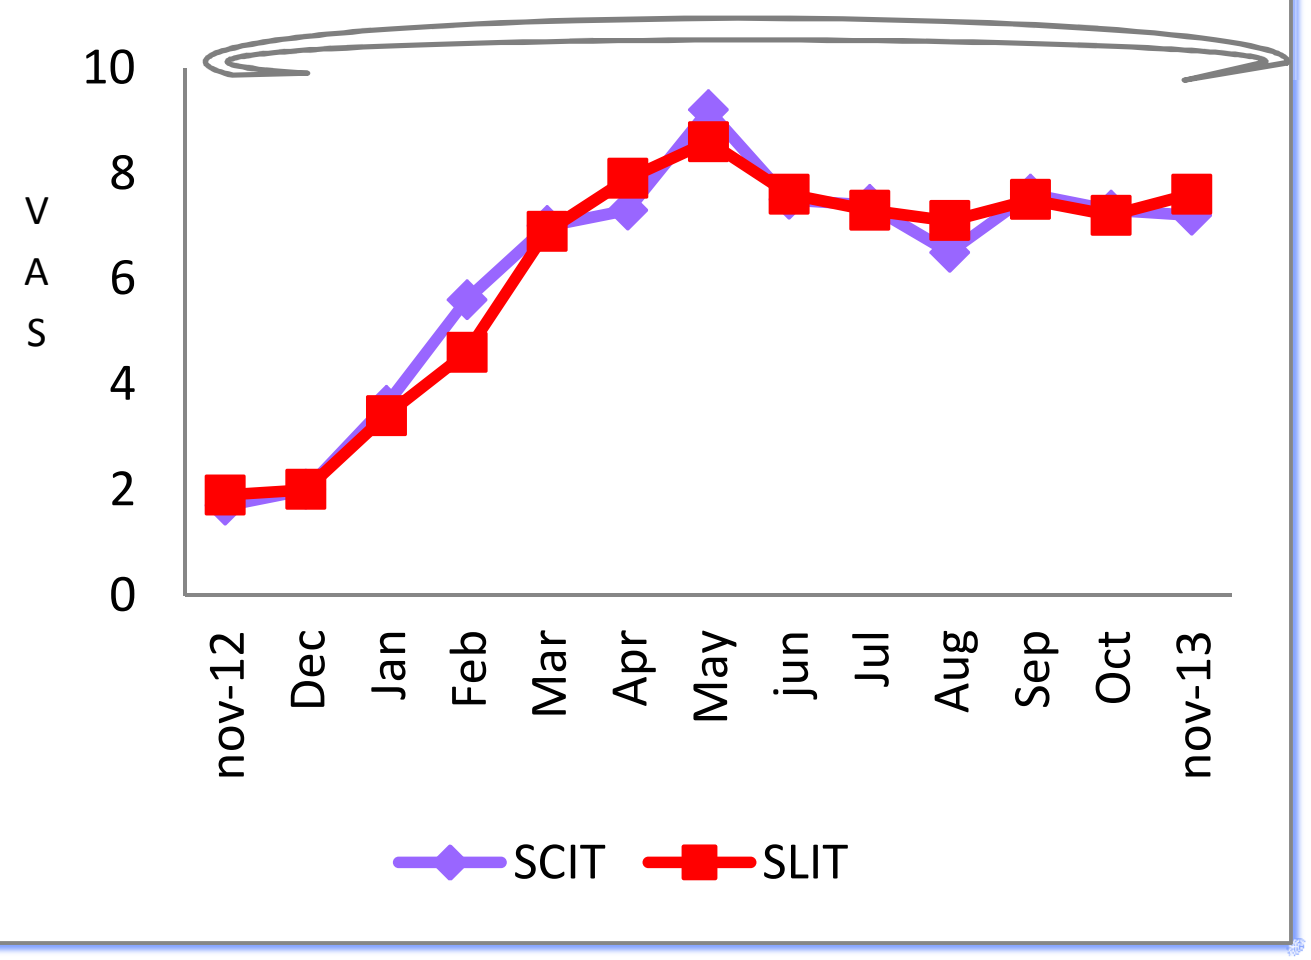
p<0.001
VAS
10
8
6
4
2
0
nov-12
Dec
Jan
Feb
Mar
Apr
May
jun
Jul
Aug
Sep
Oct
nov-13
SCIT
SLIT